498: ERBB2/HER2 Status In Biliary Tract Cancer Patients In Spanish RETUD Registry

AJ Muñoz[1], F Castet[2], R Vera[3], R Álvarez[4], C Pertusa[5], P Garcia-Alfonso[1], R Jiménez[1], R Vidal-Tocino[6], I Ales[7], N Luque[8], J Adeva[9], WG Parra[9], A Carral[10], RM Rodriguez-Alonso[11], E Aranda[11], P Cerda[12], E Martínez de Castro[13], A La Casta[14], B Laquente[15], T Macarulla[2]

[1]Medical Oncology Department, Hospital General Universitario Gregorio Marañón, Madrid, Spain; [2]Medical Oncology Department, Hospital Clínic, Barcelona, Spain; [3]Medical Oncology Department, Hospital Universitario de Navarra, Navarra, Spain; [4]Medical Oncology Department, Hospital Universitario HM Sanchinarro, Madrid, Spain; [5]Medical Oncology Department, Consorcio Hospital General Universitario de Valencia, Valencia, Spain; [6]Medical Oncology Department, Hospital Universitario de Salamanca, IBSAL, Salamanca, Spain; [7]Medical Oncology Department, Hospital Regional Universitario de Málaga, Málaga, Spain; [8]Medical Oncology Department, Hospital Universitario Virgen de las Nieves, Granada, Spain; [9]Medical Oncology Department, Hospital Universitario 12 de Octubre, Madrid, Spain; [10]Medical Oncology Department, Hospital Universitario de A Coruña, A Coruña, Spain; [11]Medical Oncology Department, Hospital Universitario Reina Sofía, Córdoba, Spain; [12]Medical Oncology Department, Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; [13]Medical Oncology Department, Hospital Universitario Marqués de Valdecilla, Santander, Spain; [14]Medical Oncology Department, Hospital Universitario de Donostia, San Sebastián, Spain; [15]Medical Oncology Department, Catalan Institut of Oncology- IDIBELL, Barcelona, Spain

GRUPO DE TRATAMIENTO DE LOS TUMORES DIGESTIVOS

REGISTRY RETUD

Background

Biliary tract cancer (BTC) is a heterogeneous group of tumors, including cholangiocarcinoma (intra and extrahepatic) and gallbladder carcinoma.[1] ERBB2 gene amplification or HER2 receptor overexpression is a well-known oncogenic driver and represents a therapeutic target in BTC.[2,3] We aimed to describe the epidemiology characteristics, treatment patterns and efficacy outcomes of a cohort of BTC patients according to HER2 expression status.

Methods

BTC diagnosis occurred between January 1st, 2017, and May 30th, 2025. Two different populations were defined for analysis **(Figure 1). P**atients initially with resectable disease (RD) and tumor relapse were analyzed in both groups.

Figure 1. Definition of patient populations for the analysis

Resectable disease population (RD)

- ✓ Patients with neoadjuvant therapy, radical surgery or locoregional therapy as first therapeutic approach

Advanced disease population (AD)

- ✓ Patients from RD population with tumor relapse
- ✓ First line systemic treatment as first therapeutic approach

ERBB2/HER2 status was assessed by next-generation sequencing (NGS), immunohistochemistry (IHC), and/or fluorescence/chromogenic in situ hybridization (FISH/CISH). HER2+ status was considered when it was overexpressed (IHQ+++) and/or when it was amplified by FISH/CISH (if IHQ++) or NGS.
Analysis comprised demographic and clinical data, tumor molecular profile, therapeutic procedures and efficacy outcomes. Time to next treatment (TTNT), progression-free survival (PFS) and overall survival (OS) were estimated using the Kaplan-Meier method. P-value < 0.05 was considered statistically significant. All analysis were done using statistical software R version 4.5.1.

Results

ERBB2/HER2 status was determined in 448 BTC patients from 31 centers. At diagnosis, 148 patients (33.0%) presented RD and 300 (67.0%) presented AD. 128 patients from RD population experienced tumor relapse.
HER2+ was confirmed in 24 (5.4%) patients, main baseline and clinic characteristics are shown in **table 1** and **table 2**.

Table 1. Demographic and clinic characteristics by HER2 status

Variable	HER2 positive N = 24	HER2 negative N = 424	p-value
Median age at diagnosis (Q1, Q3)	67.50 (56.00, 72.50)	66.00 (59.00, 73.00)	0.857
Sex (female)	18 (75.00%)	186 (43.87%)	**0.003**
Median BMI (Q1, Q3)	25.65 (22.31, 29.70)	25.95 (23.37, 28.72)	0.726
Median follow-up time (Q1, Q3)	16.32 (11.35, 23.42)	16.61 (9.77, 26.61)	0.771
Advanced disease	24 (100.00%)	404 (95.28%)	0.616
Resectable disease	7 (29.17%)	141 (33.25%)	0.679
ECOG			0.191
0	3 (14.29%)	100 (30.30%)	
1	11 (52.38%)	169 (51.21%)	
2	2 (9.52%)	26 (7.88%)	
3	0 (0.00%)	3 (0.91%)	
Unknown	5 (23.81%)	32 (9.70%)	
Missing	3	94	

Table 2. HER2 status by tumor location

Variable	Ampulla of Vater	Extrahepatic distal	Extrahepatic hiliar	Gallbladder
HER2 amplification				
HER2 positive	0 (0.00%)	2 (3.28%)	3 (7.32%)	9 (15.25%)
HER2 negative	9 (100.00%)	59 (96.72%)	38 (92.68%)	50 (84.75%)
Variable	**Intra-extrahepatic**	**Intrahepatic**	**Unknown**	**p-value**
HER2 amplification				**0.002**
HER2 positive	0 (0.00%)	9 (3.26%)	1 (100.00%)	
HER2 negative	1 (100.00%)	267 (96.74%)	0 (0.00%)	

Treatments received by RD and AD populations are shown in **table 3** and **table 4,** respectively.

Table 3. Treatments for resectable disease population

Variable	HER2 positive N = 7	HER2 negative N = 141	p-value
Surgery[1]	7 (100.00%)	130 (92.20%)	1.000
Hepatectomy	4 (57.14%)	74 (56.92%)	1.000
Lymphadenectomy	3 (42.86%)	35 (26.92%)	0.396
Duodeno-pancreatectomy	1 (14.29%)	42 (32.31%)	0.433
Cholecystectomy	2 (28.57%)	21 (16.15%)	0.334
Neoadjuvant treatment	2 (28.57%)	12 (8.51%)	0.171
Adjuvant treatment	4 (57.14%)	83 (58.87%)	1.000
ORR for neoadjuvant treatment	0 (0.00%)	4 (50.00%)	0.467
Not evaluable	0	4	

[1]One patient can have more than one procedure

Table 4. Treatments for advanced disease population

Variable	HER2 positive N = 24	HER2 negative N = 404	p-value
Surgery[1]	1 (4.17%)	5 (1.24%)	0.294
Hepatectomy	1 (100.00%)	4 (80.00%)	1.000
Lymphadenectomy	1 (100.00%)	1 (20.00%)	0.333
Cholecystectomy	1 (100.00%)	2 (40.00%)	1.000
First line	24 (100.00%)	399 (98.76%)	1.000
ORR for first line treatment	7 (36.84%)	95 (28.88%)	0.458
Not evaluable	5	70	
Second line	17 (70.83%)	226 (55.94%)	0.152
ORR for second line treatment	1 (7.69%)	21 (11.67%)	1.000
Not evaluable	4	46	

[1]One patient can have more than one procedure

A total of 7 **(29.2%)** HER2+ patients received **anti-HER2** therapy and 3 (**12.5%)** received **immunotherapy.**

Median OS (mOS) in **HER2+** AD patients treated with **anti-HER2 therapies** was 32.7 months (95% CI 20.7-NA); mOS for HER2+ AD patients with other treatments was 13.1 months (95% CI 9.3-17.0) **(p=0.003) (Figure 2a)** versus mOS in **HER2- AD** patients of 15.0 months (95% CI 13.5-17.3) **(p=0.012) (Figure 2b).**

Figure 2. Overall survival. **a)** HER2+ patients receiving anti-HER2 therapy vs HER2+ patients without anti-HER2 therapy. **b)** HER2+ patients without anti-HER2 therapy vs HER2- patients

a)

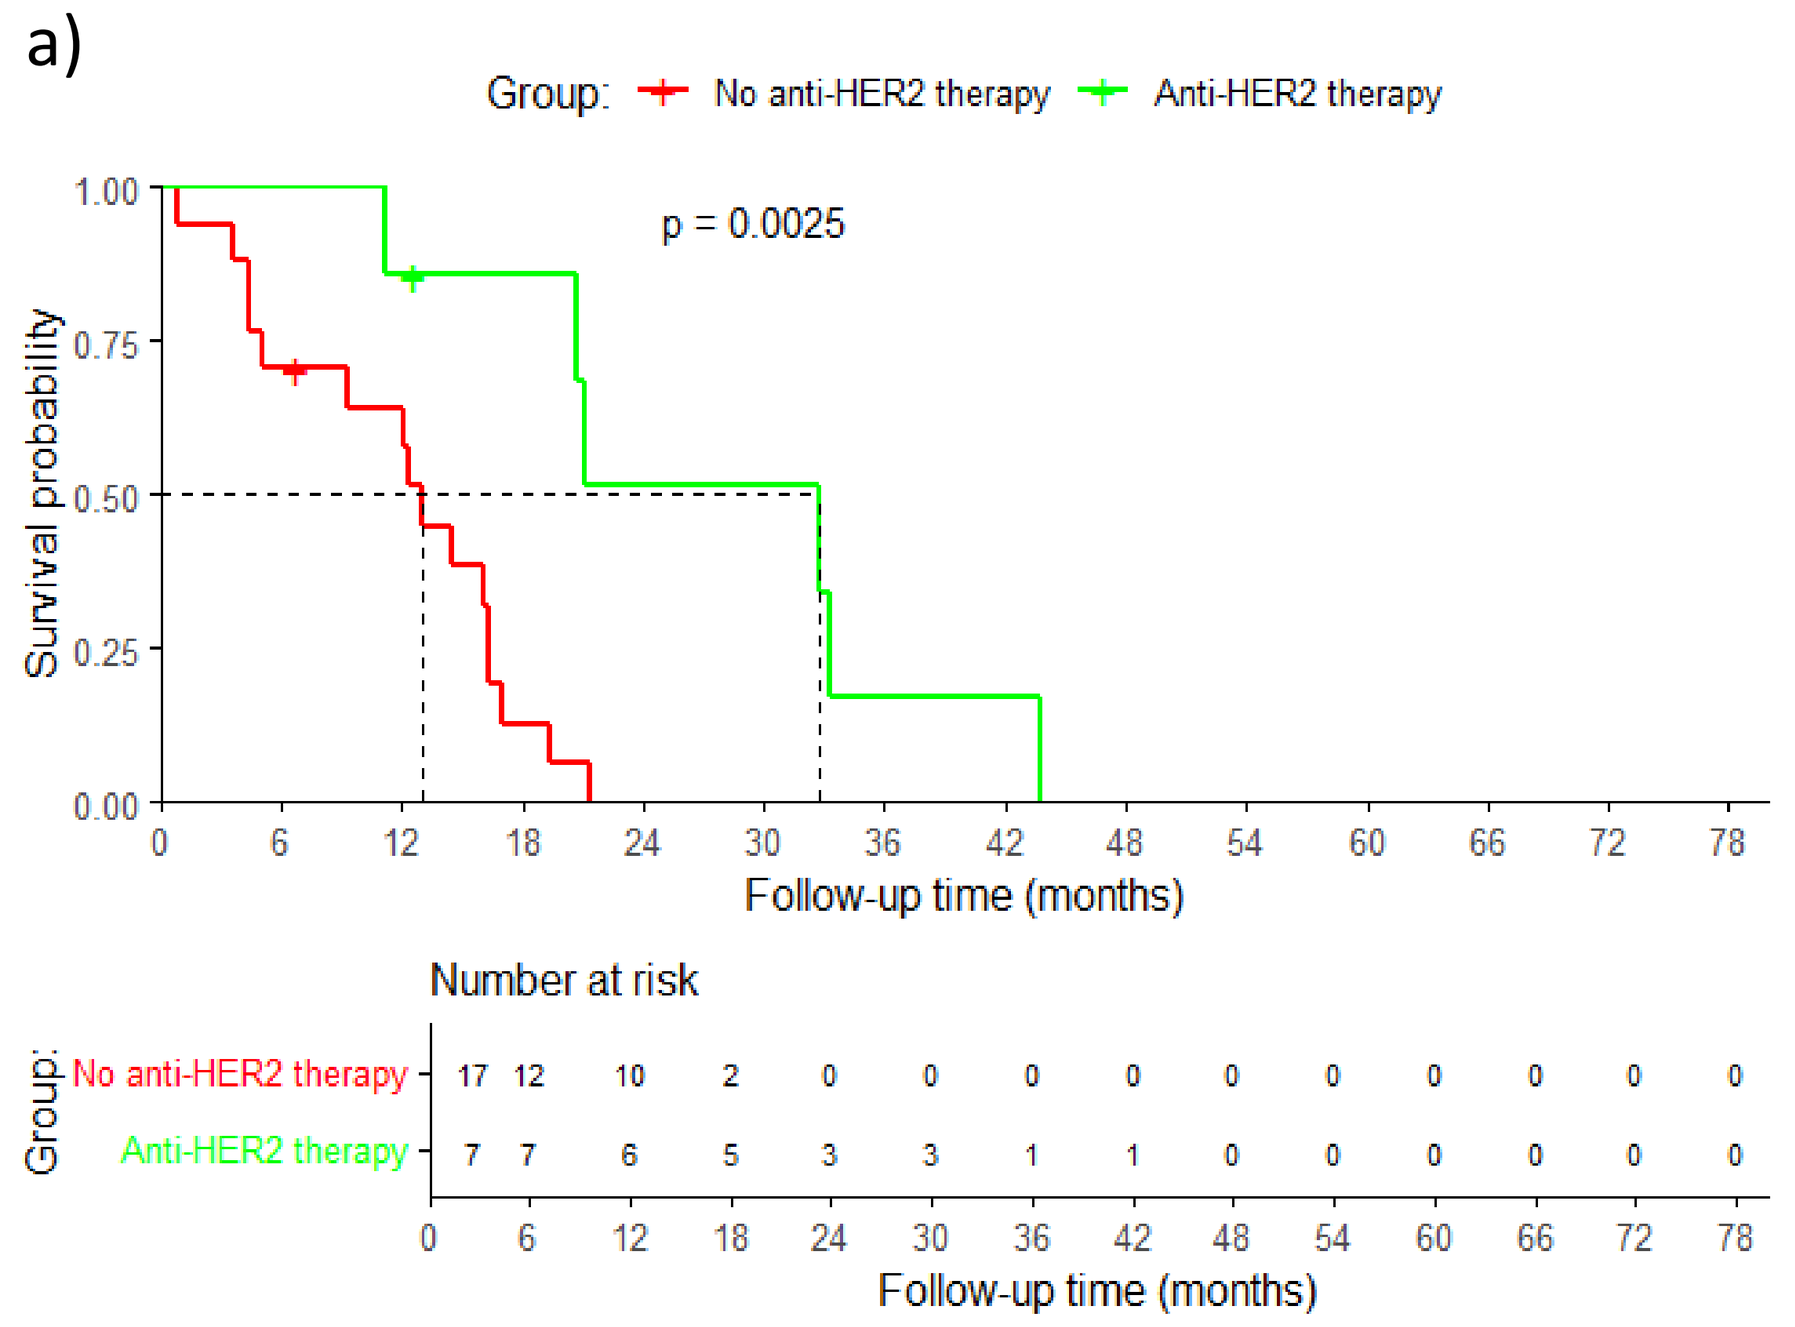
Group: No anti-HER2 therapy Anti-HER2 therapy
p = 0.0025
Survival probability
Follow-up time (months)
Number at risk
No anti-HER2 therapy 17 12 10 2 0 0 0 0 0 0 0 0 0 0
Anti-HER2 therapy 7 7 6 5 3 3 1 1 0 0 0 0 0 0
Group
Follow-up time (months)

b)

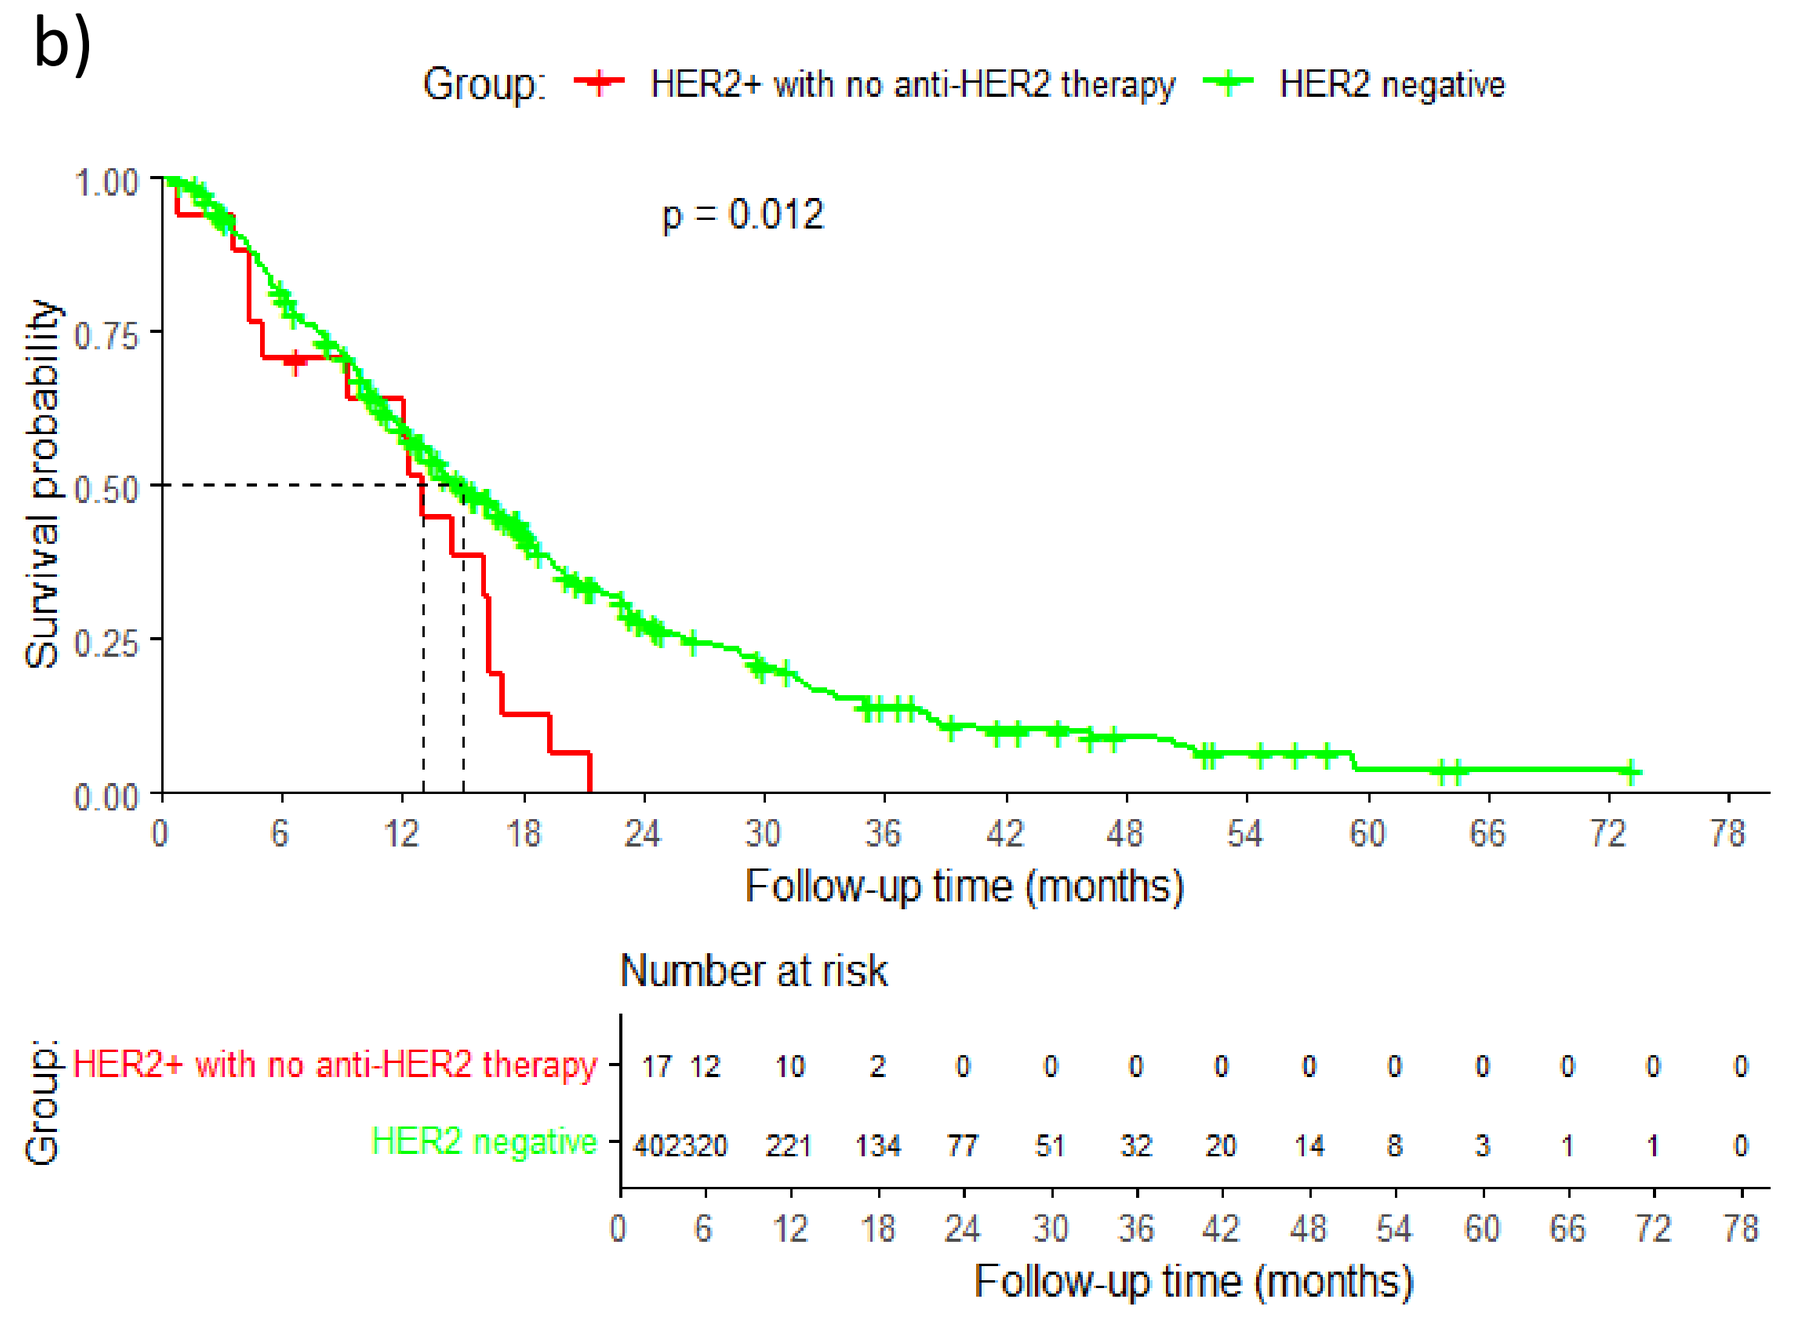
Group: HER2+ with no anti-HER2 therapy HER2 negative
p = 0.012
Survival probability
Follow-up time (months)
Number at risk
HER2+ with no anti-HER2 therapy 17 12 10 2 0 0 0 0 0 0 0 0 0 0
HER2 negative 402320 221 134 77 51 32 20 14 8 3 1 1 0
Group
Follow-up time (months)

For HER2+ patients mPFS in first line was 6.6 months (95% CI 4.5-8.1) and 6.6 months (95% CI 5.9-7.7) for HER2- (p=0.075). HER2+ patients presented a mTTNT of 7.8 months (95% CI 6.7-NA) vs 10.0 months (95% CI 9.2-11.2) in HER2- patients (p=0.047).

This analysis provides insights into the **characterization** of real-world **HER2+ BTC** patients, with **anti-HER2 therapies** showing a significant impact in **mOS**

Acknowledgements

The authors want to thank the patients and the staff of all the participating institutions. The RETUD Registry has been funded by the TTD, through an unrestricted grant from AstraZeneca Farmacéutica Spain S.A, Incyte Biosciences International Sàrl, Laboratorios Servier S.L. and other pharmaceutical companies.

Contact information

Email: andresmunmar@hotmail.com

References

1. Gray S et al. Cancers. 2022;14(7):17892. **2.** Lee SS et al. Clin Cancer Res. 2025 Nov 14 **3.** Slamon DJ et al. N Engl J Med. 2001 Mar 15;344(11):783-92.